Ofranergene Obadenovec (VB-111) in Platinum-Resistant Ovarian Cancer: Favorable Response Rates in a Phase I/II Study are Associated with an Immunotherapeutic Effect

Authors: Yael C. Cohen MD[1], Suzanne T. Berlin DO[2], Michael J. Birrer MD PhD[3], Susana M. Campos MD[4], Tamar Rachmilewitz Minei MD[1], Dror Harats MD[1], Richard T. Penson MD MRCP[3]

Affiliations: *[1]VBL Therapeutics, Modi'in, Israel; [2]Dana–Farber Cancer Institute, York, ME; [3]Massachusetts General Hospital, Harvard Medical School, Boston, MA; [4]Dana-Farber Cancer Institute, Boston, MA*

BACKGROUND

VB-111 is a Novel, First-in-class Anti-cancer Gene Therapy

- VB-111 (ofranergene obadenovec) is a targeted anti-cancer viral gene therapy, based on a non-replicating non-integrating Adenovirus 5 vector that contains a transgene targeting angiogenic blood vessels
- VB-111 has a dual mechanism of action:
 - Apoptosis of angiogenic endothelium and vascular disruption, leading to tumor starvation
 - Induction of an anti-tumor directed immune response
- Administered by IV infusion once every 8 weeks
- Well tolerated in over 300 cancer patients
- We report final results of a Phase I/II study of VB-111 in combination with paclitaxel in patients with platinum-resistant ovarian cancer

Ovarian Cancer

- Ovarian cancer is the leading cause of death from gynecologic cancers; Disease is difficult to detect and often diagnosed only when advanced
- Most patients ultimately develop platinum-resistant relapse
- Anti-angiogenic therapy has been shown to be efficacious in ovarian cancer. Bevacizumab was recently shown to improve Progression Free Survival (PFS) (but not Overall Survival (OS)) in combination with chemotherapy, in platinum-resistant recurrent ovarian cancer (AURELIA trial*)

STUDY DESIGN

- Study NCT01711970 was a prospective, open label, dose escalating Phase I/II study assessing combination treatment of VB-111 Q8W and weekly paclitaxel
- **Population:** Patients with recurrent platinum-resistant ovarian cancer
- **Treatment:**
 - Phase I: escalating doses of intravenous VB-111 and paclitaxel to identify dose limiting toxicities
 - Phase 2: therapeutic doses of VB-111 1x10^{13} Viral Particles and paclitaxel 80mg/m2 until disease progression
- **Assessments:** Safety, OS, PFS, tumor response (CA-125 and RECIST 1.1) and histopathology.
- **Study objectives:**
 - Safety and tolerability of combination of VB-111 and weekly paclitaxel
 - Explorative efficacy (RECIST 1.1 response, CA-125 response, PFS and OS) in an expanded cohort of the optimally tolerated dose

* J Clin Oncol. 2014;32(13):1302-8

RESULTS

Patient Characteristics and Treatment Exposure

- 21 patients with recurrent platinum-resistant ovarian cancer were enrolled at Massachusetts General Hospital and Dana Farber Cancer Institute, Boston, MA
- All patients had measurable disease
- Enrolled patients had unfavorable prognosis factors with **~50% platinum refractory disease and 50% previous treatment with anti-angiogenics** (Table 1)
- Patients were treated in 2 Treatment Groups: Sub-Therapeutic Dose and Therapeutic Dose Intra-patient dose escalation was allowed
- Patients received a median of 2 VB-111 doses (range: 1-7)

Sub-Therapeutic Dose
N=4
VB-111 3x10^{12} VPs
+
Paclitaxel 40-80mg/m^2

Therapeutic Dose
N=17
VB-111 1x10^{13} VPs
+
Paclitaxel 80 mg/m^2

Table 1 - Baseline Characteristics

	Sub-Therapeutic Dose (N=4)	Therapeutic Dose (N=17)	All Doses (N=21)
Age, mean (years)	66.3	64.5	64.9
Race: White, n (%)	4 (100)	17 (100)	21 (100)
Histology, n (%)			
Papillary serous	1 (25.0)	8 (47.1)	9 (42.9)
Clear Cell	1 (25.0)	1 (5.9)	2 (9.5)
Adenocarcinoma	2 (50.0)	2 (11.8)	4 (19.0)
Carcinosarcoma	0	3 (17.6)	3 (14.3)
Other	0	3 (17.6)	3 (14.3)
Cancer Stage at Diagnosis, n (%)			
IA/B/C	1 (25.0)	2 (11.8)	3 (14.3)
IIIC	1 (25.0)	11 (64.7)	12 (57.1)
IV	2 (50.0)	4 (23.5)	6 (28.6)
Prior Lines of Therapy, mean {SD]	2.3 [96]	2.7 [92]	2.6 [92]
Prior Anti-Angiogenic treatment=Yes, n (%)	**3 (75.0)**	**8 (47.1)**	**11 (52.4)**
Platinum Free Interval, n (%)			
>=3 months (platinum resistant)	3 (75.0)	8 (47.1)	11 (52.4)
< 3 months **(platinum refractory)**	**1 (25.0)**	**9 (52.9)**	**10 (47.6)**
ECOG Performance status, n (%)			
00 - Fully active	2 (50.0)	8 (47.1)	10 (47.6)
01 - Restricted	2 (50.0)	9 (52.9)	11 (52.4)

Significant Increase in Overall Survival

Median OS: 498 Vs. 172.5 days Therapeutic dose (n=17) vs. Low dose (n=4)

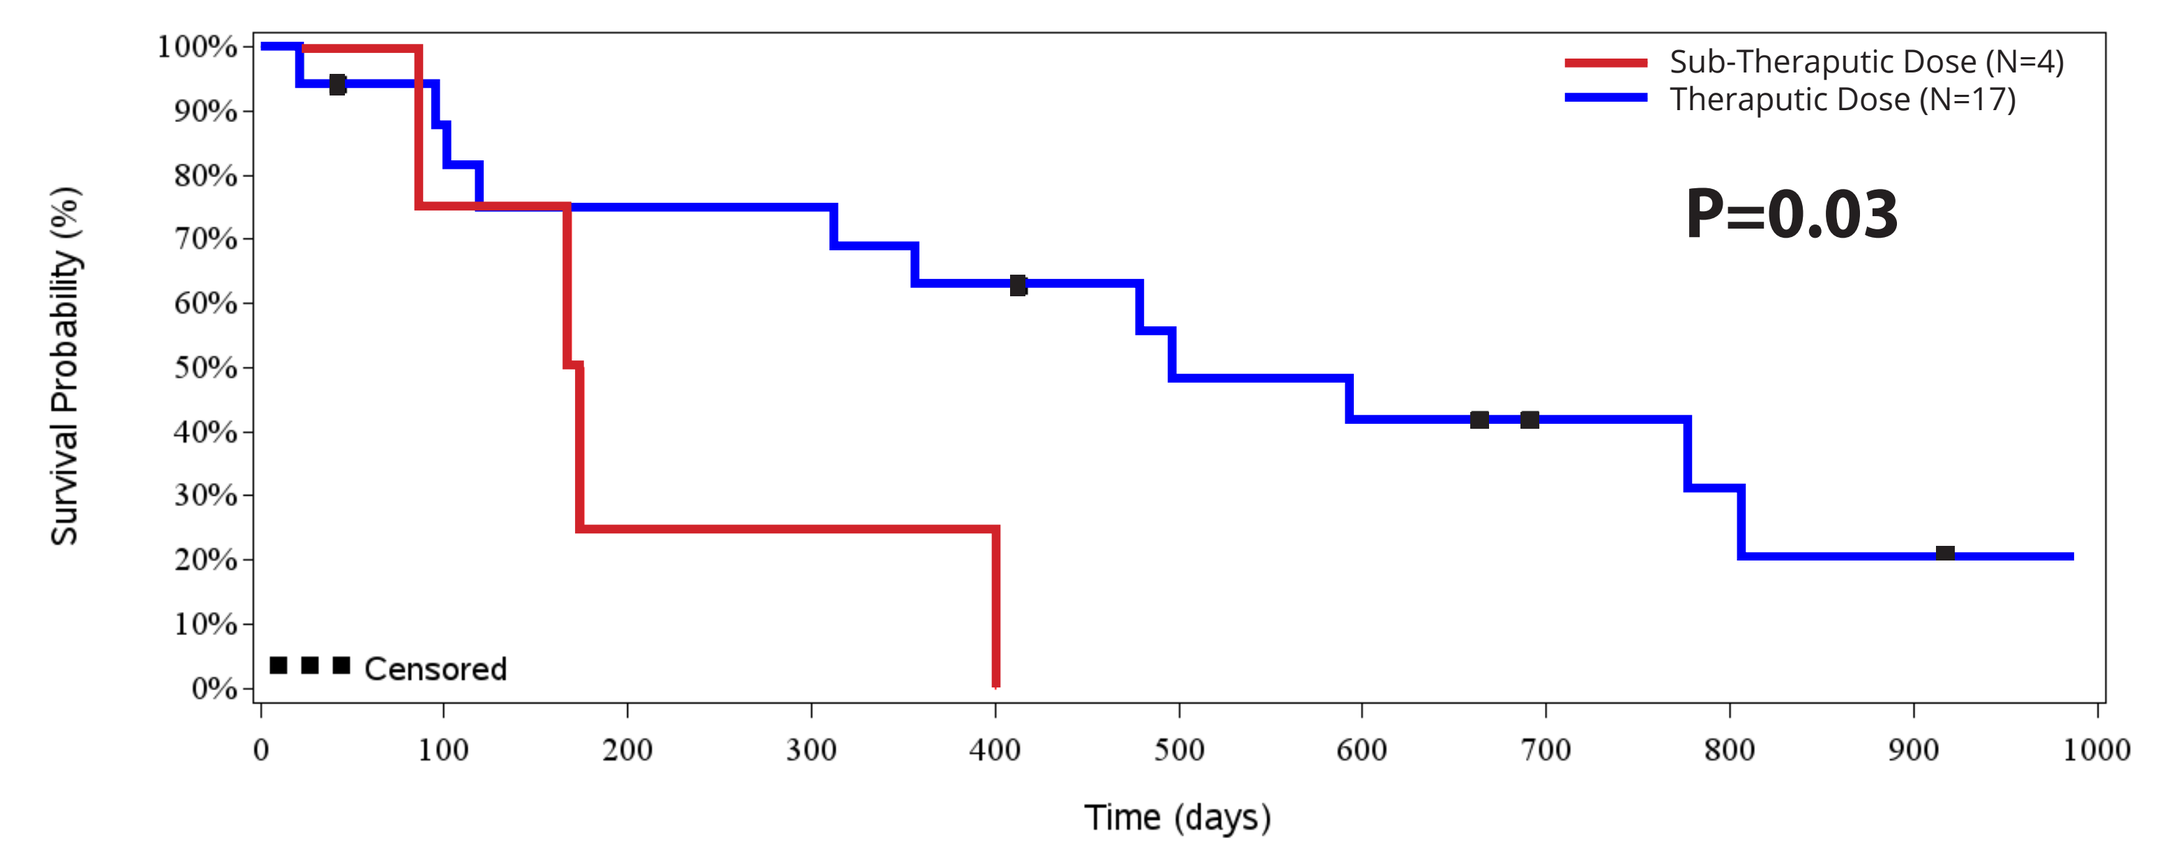
Sub-Theraputic Dose (N=4)
Theraputic Dose (N=17)
P=0.03
Censored
Survival Probability (%)
Time (days)

Durable Disease Control: CA-125 Change

Individual Percent Change in CA-125 Level

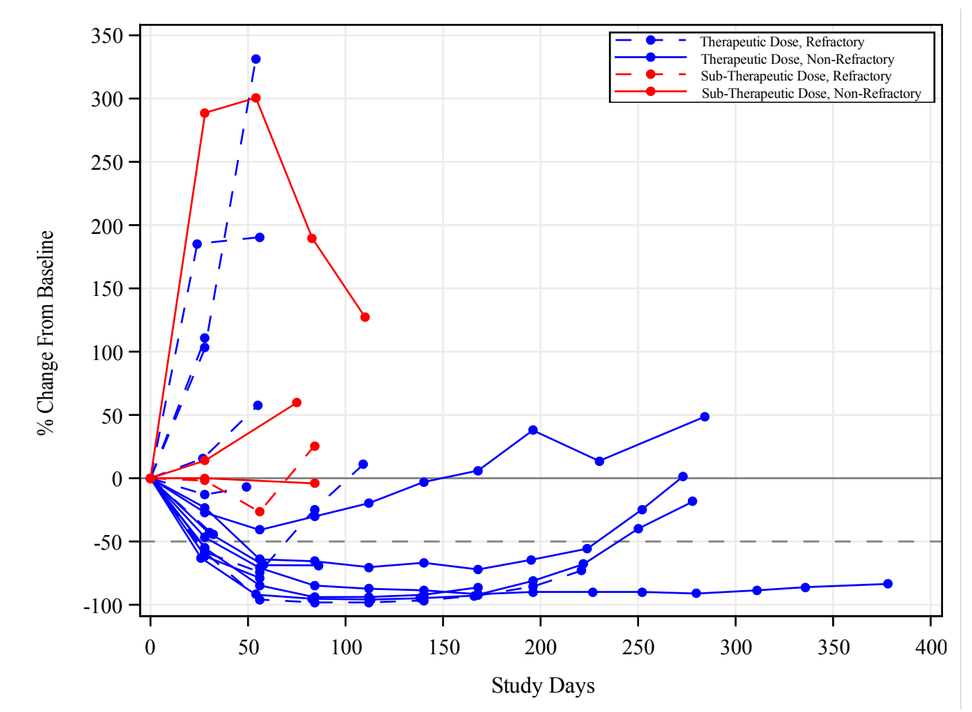
% Change From Baseline
Study Days

Durable Disease Control: Tumor Burden Change

Individual Percent Change in Tumor Burden (Sum of LD)

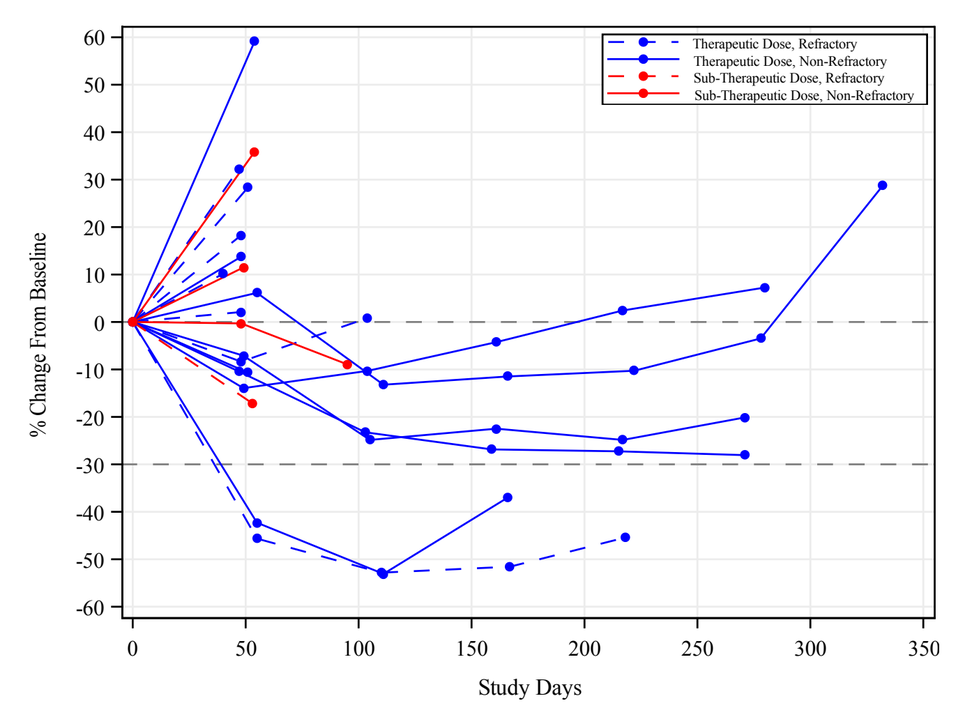
% Change From Baseline
Study Days

CA-125 or Fever are Predictive Biomarkers for OS Benefit in Ovarian Cancer Patients Treated with VB-111

Post treatment fever (reported by 29% treated with Therapeutic Dose) was associated with a signal for improved survival: mOS of 808 days in patients with fever compared to 479 days in patients without fever (p=0.27)

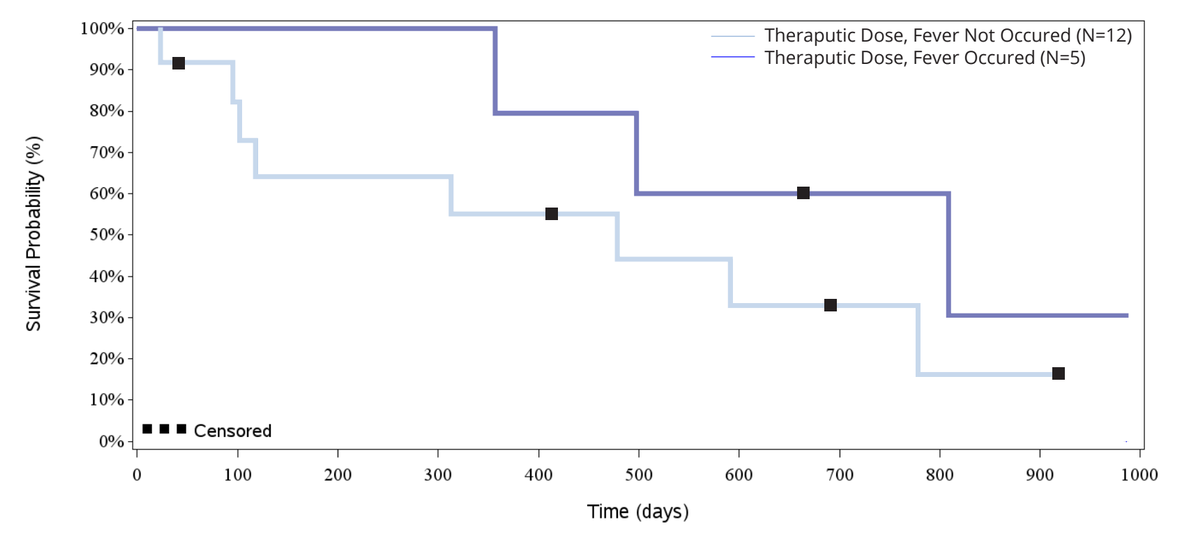
Theraputic Dose, Fever Not Occured (N=12)
Theraputic Dose, Fever Occured (N=5)
Censored
Survival Probability (%)
Time (days)

CA-125 decrease was associated with a signal for improved survival. Median OS was 808 vs. 351 days in patients with CA-125 decrease of 50% compared to those without 50% decrease in CA-125 (P=0.067)

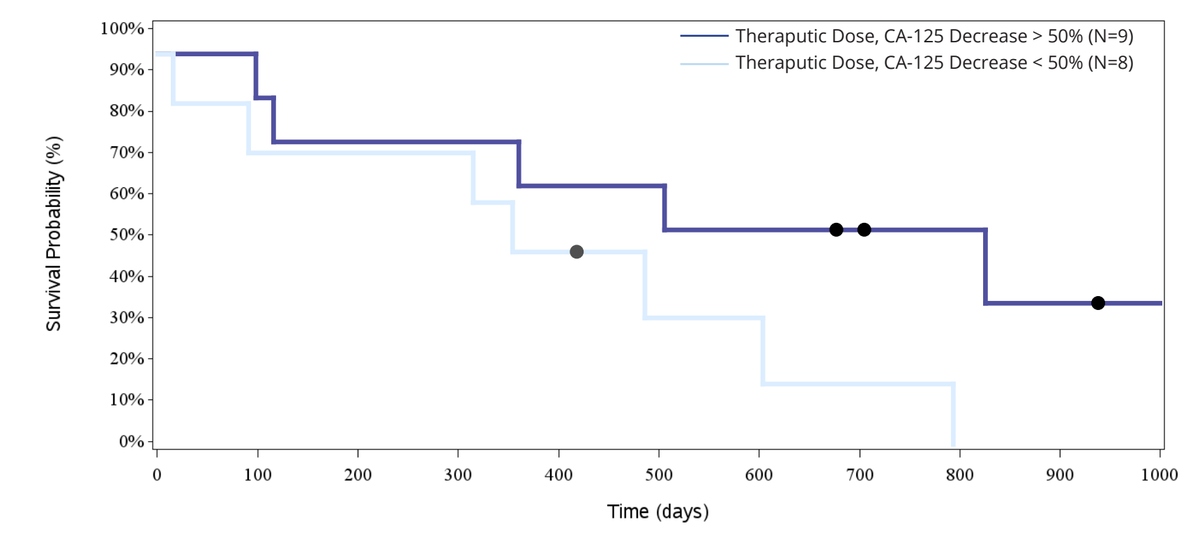
Theraputic Dose, CA-125 Decrease > 50% (N=9)
Theraputic Dose, CA-125 Decrease < 50% (N=8)
Survival Probability (%)
Time (days)

GCIG CA-125 Response Rate of 58% Doubled Compared to AURELIA Historical Data

58.3% (7/12) of evaluable patients treated with the therapeutic dose had a GCIG CA-125 response (defined as 50% reduction in CA-125 confirmed over 4 weeks), compared with 31.8% with bevacizumab and chemotherapy, and 11.6% with chemotherapy alone in the AURELIA trial*

Individual CA-125 Maximum Decrease by Treatment

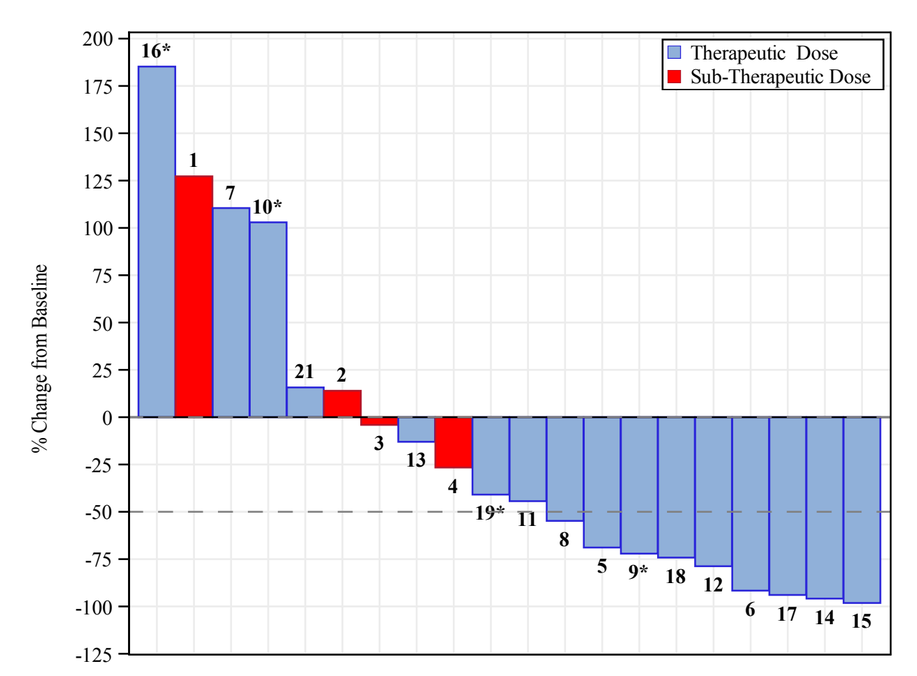
Therapeutic Dose
Sub-Therapeutic Dose
% Change from Baseline

Tumors Transformed from Immunologically "Cold" to "Hot"

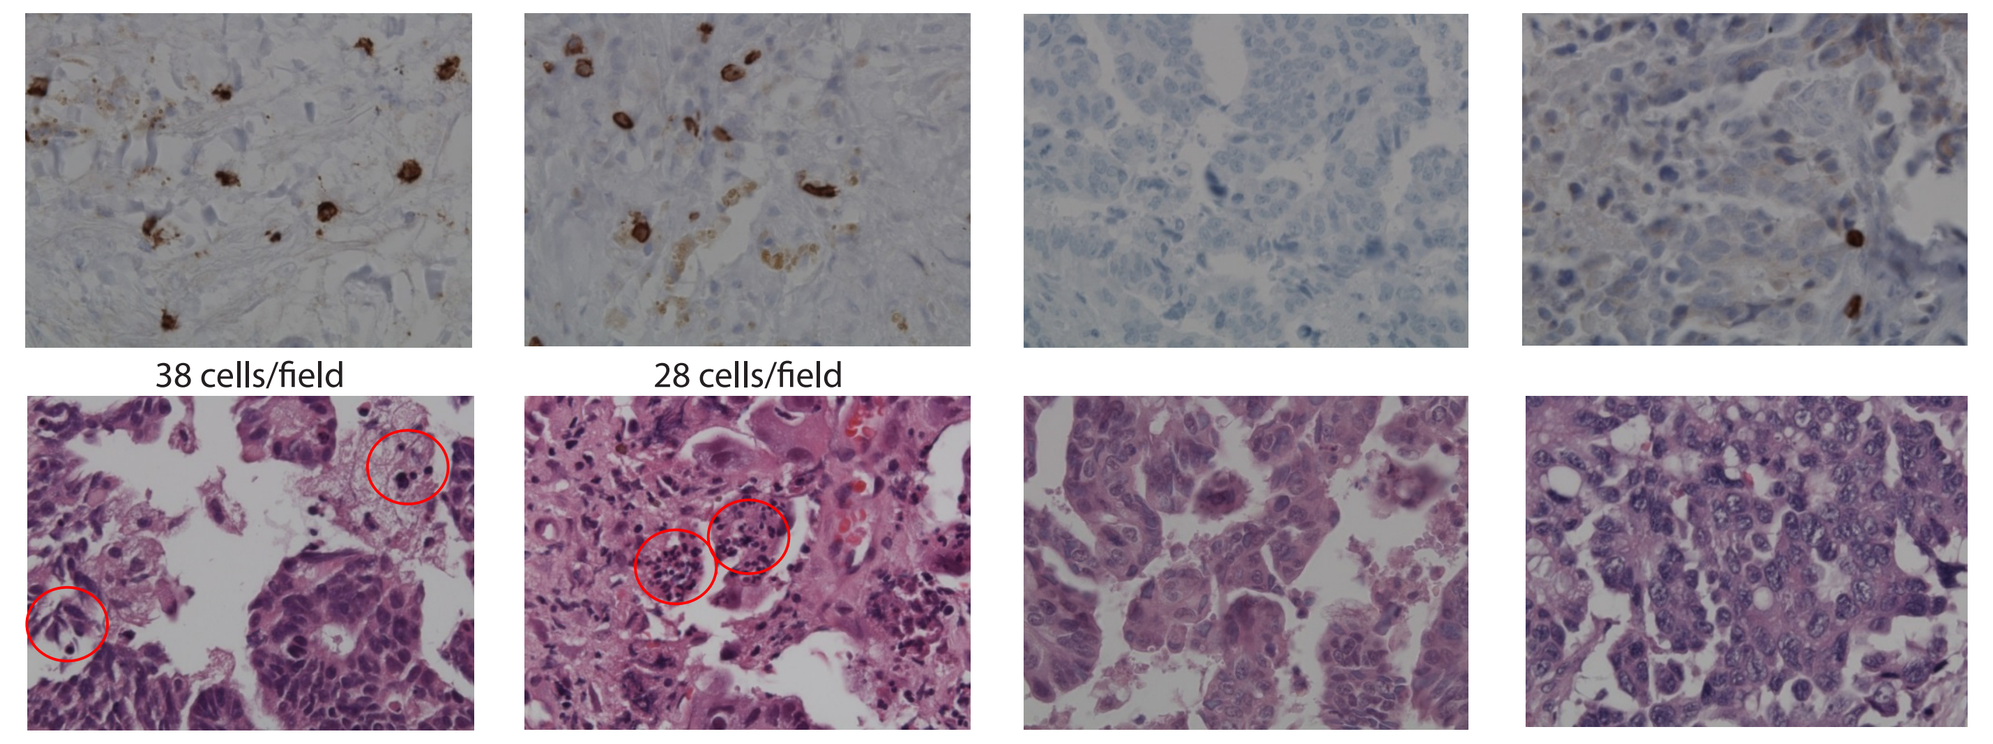
VB-111 Pt 1
VB-111 Pt 2
Control Pt 1
Control Pt 2
CD8
(x400)
38 cells/field
28 cells/field
H&E
(x400)

Biopsies were obtained from patients with Recurrent Platinum-Resistant Ovarian Cancer. H&E and Immunohistochemistry staining show regions of apoptotic tumor cells (red circles) and increased tumor infiltrating CD8 lymphocytes in VB-111 treated patients. Specimens from untreated controls showed minimal or no tumor T cell infiltration

Safety

- VB-111 was well tolerated other than fever and the anticipated side-effects of an anti-angiogenic
- Toxicity on the combination was similar to the expected with anti-angiogenics and taxanes in this patient population
- One toxicity causing treatment discontinuation was reported: an anaphylaxis reaction confirmed after re-challenge to be related to paclitaxel
- 9 patients (43%) reported AEs grade >=3
- Most frequent AEs included: fatigue (52%), nausea (52%), fever (48%), anemia (38%), diarrhea (33%), headache (29%)
- Most common VB-111 related AEs were transient mild-moderate fever/flu like symptoms

SUMMARY AND CONCLUSIONS

- **Safety:** VB-111 in combination with paclitaxel, was safe and well tolerated
- **Efficacy signal** was seen in patients treated with the therapeutic dose despite unfavorable prognostic characteristics:
 - **CA-125 responses (GCIG)** reported in **58%** of evaluable patients
 - **Survival:** significant increase in OS at therapeutic vs. sub therapeutic dose **498 vs. 172.5 days, p=0.03**
 - **Biomarkers:** A trend for improved survival seen in patients with post treatment fever and in patients with a 50% decrease in CA-125
 - **Immunotherapeutic effect:** post treatment tumor infiltrating CD8 T-cells and apoptotic cancer cells indicating tumor transformation from immunologically 'cold' to immunologically 'hot', possibly contributing to the favorable clinical outcomes
- Encouraging results are the basis for further exploration in the randomized controlled, pivotal OVAL study, NCT03398655 that is currently recruiting patients with platinum-resistant ovarian cancer